Improving CsCl Density Gradient Ultracentrifugation Process Through Automated Gradient Formation: Evaluation of the OptiMATE Gradient Maker

FORGE BIOLOGICS
A member of Ajinomoto Bio-Pharma Services

BECKMAN COULTER Life Sciences

Anto Nirmal Louis Babu[a], Smita Menon[a], Aghiad Alkhiamy[a], Ganesh Krishnamoorthy[a], Frank Agbogbo[a], Aparna Jasti[b], Shawn Sternisha[b] , Balasubramanian Venkatakrishnan[b] and David Dismuke[a]

[a]Forge Biologics, 3900 Gantz Drive, Grove City, OH 43123

[b]Beckman Coulter Life Sciences, 5350 Lakeview Parkway S. Drive, Indianapolis, IN 46268

ABSTRACT

Recombinant adeno-associated virus (rAAV) is a popular choice as a gene therapy delivery vector due to its sustained transient expression levels, safety and low immunogenicity. It is used in the treatment of numerous genetic disorders with more than 225 conducted clinical trials and 6 FDA approved gene therapy products.[1]

Forge Biologics utilizes cesium chloride (CsCl) density gradient ultracentrifugation as a platform process step to enrich full AAV vector capsids. CsCl density gradient ultracentrifugation is a gold-standard for the separation of the full/empty capsids. Although the process is commonly used as an industry standard, scale-up, processing times, consistency and reproducibility are some critical challenges associated with the use of this technology. Key bottlenecks include manual filling, precise tube balancing, longer spin times, and inconsistent manual tube sealing that can impact process consistency and reproducibility.

To address these challenges, in collaboration with the Beckman Coulter Life Sciences we evaluated the use of a pre-formed continuous CsCl gradient using the OptiMATE Gradient Maker (Beckman Coulter Life Sciences) and compared it to our established CsCl isopycnic density gradient process with respect to processing time, recovery and product quality. The use of a pre-formed gradient significantly shortens the process time while maintaining resolution and improving process throughput. The separated full capsids were recovered using the OptiXTRACT sample recovery tool (Beckman Coulter Life Sciences) and tested for recovery efficiency and purity. We found that the recovery efficiency of full AAV capsids was similar >75% in both cases. The purity of the full capsids was approximately 88% in both cases as analyzed by analytical ultracentrifugation.

Given the high production costs for AAV, it is important to minimize process times and reduce operator-dependent variability. Overall, the results indicate that the automated gradient maker performs comparably to our established CsCl platform in both recovery and product quality, while offering significant time savings and reproducibility for large-scale operations.

AAV DOWNSTREAM PURIFICATION PROCESS FLOW

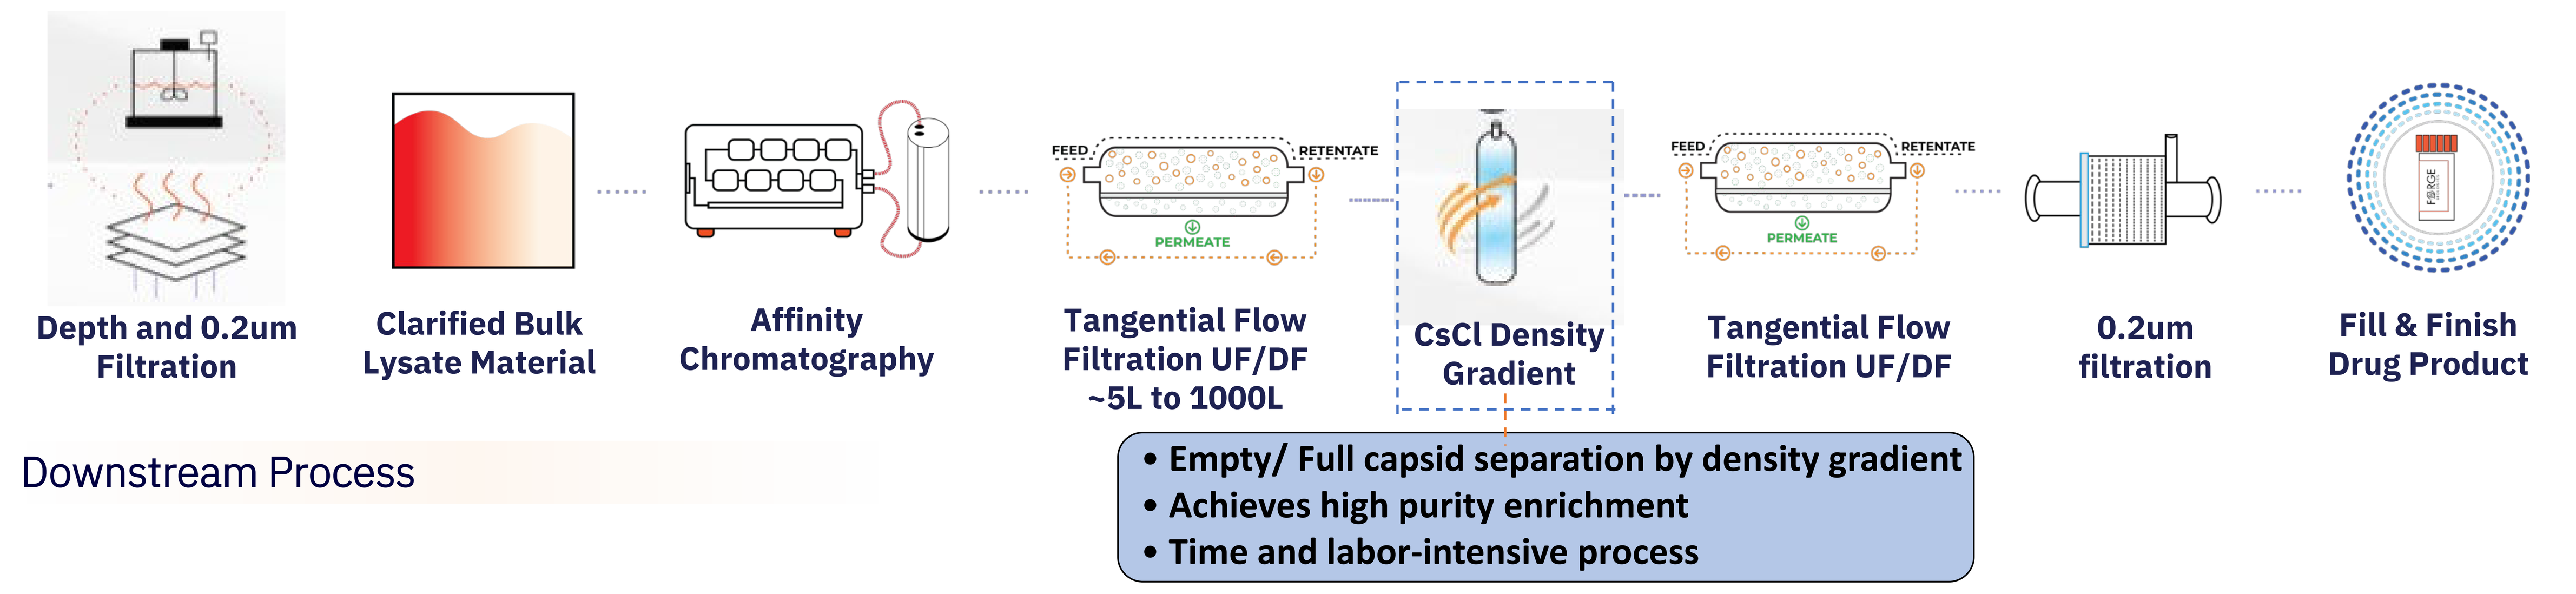
Depth and 0.2um Filtration
Clarified Bulk Lysate Material
Affinity Chromatography
FEED
RETENTATE
PERMEATE
Tangential Flow Filtration UF/DF ~5L to 1000L
CsCl Density Gradient
Tangential Flow Filtration UF/DF
0.2um filtration
Fill & Finish Drug Product
Downstream Process
• Empty/ Full capsid separation by density gradient
• Achieves high purity enrichment
• Time and labor-intensive process

Figure 1. AAV Purification Workflow. The process includes clarification, affinity capture, concentration and buffer exchange using TFF, CsCl density gradient empty/full capsid separation, final formulation TFF, final filtration, and fill-finish to produce the Drug Product.

CURRENT CsCl PROCESS CHALLENGES

- **Manual Filling**: Trapped air bubbles during manual filling can deform ultracentrifugation tubes and disturb CsCl gradients, reducing separation quality.
- **Balancing Tubes**: Precise balancing of tubes is required to maintain integrity during high-speed centrifugation which is tedious, time-consuming, and prone to operator error process.
- **Long Spin Times**: Self gradient ultracentrifugation runs often exceed 16 hours, extending the total process times to ~2-3 days which significantly limits manufacturing throughput.
- **Variability during Sealing**: Manual tube sealing can lead to improper closures, causing leaks, material loss, compromising tube integrity, and increasing process failure risk.
- **Operator- to- Operator Variability**: Multiple manual steps introduce operator-to-operator variability, making standardization difficult and reducing process reproducibility in manufacturing.

OptiMATE Gradient Maker FEATURES

- **Automated Gradient Formation & Pre-Formed Linear Gradients**: Enables precise, reproducible stepwise and linear gradient generation while eliminating manual variability. Pre-formed gradients significantly shorten ultracentrifugation spin times, improving throughput.
- **Automated Tube Sealing & Reduced Operator Burden:** Standardizes tube sealing to minimize leakage and ensure integrity, while reducing manual steps such as filling and balancing. This lowers operator workload and variability across runs.
- **Reagent Flexibility**: Compatible with multiple separation media including Cesium Chloride (CsCl), Iodixanol (IDX), and sucrose.
- **Ease of Tech Transfer**: Backed by published methods, enabling smooth transition of processes from development to GMP-scale manufacturing.
- **Enhanced Process Robustness**: Combining automation, reduced spin times, and standardized handling strengthens scalability, safety, and reproducibility of AAV full/empty capsid separations.

OptiMATE SYSTEM AND TRADITIONAL PROCESS SETUP

- **OptiMATE System Process**: User defined gradient parameters, such as upper and lower limit range for density, gradient type (linear or stepwise), and total tube volume. The OptiMATE system is programmed to automatically fill the tubes and seal them.
- **Traditional CsCl Process (Forge Platform)**: Includes Manual mixing of CsCl with AAV to target a specific refractive index and manual tube fill and sealing.

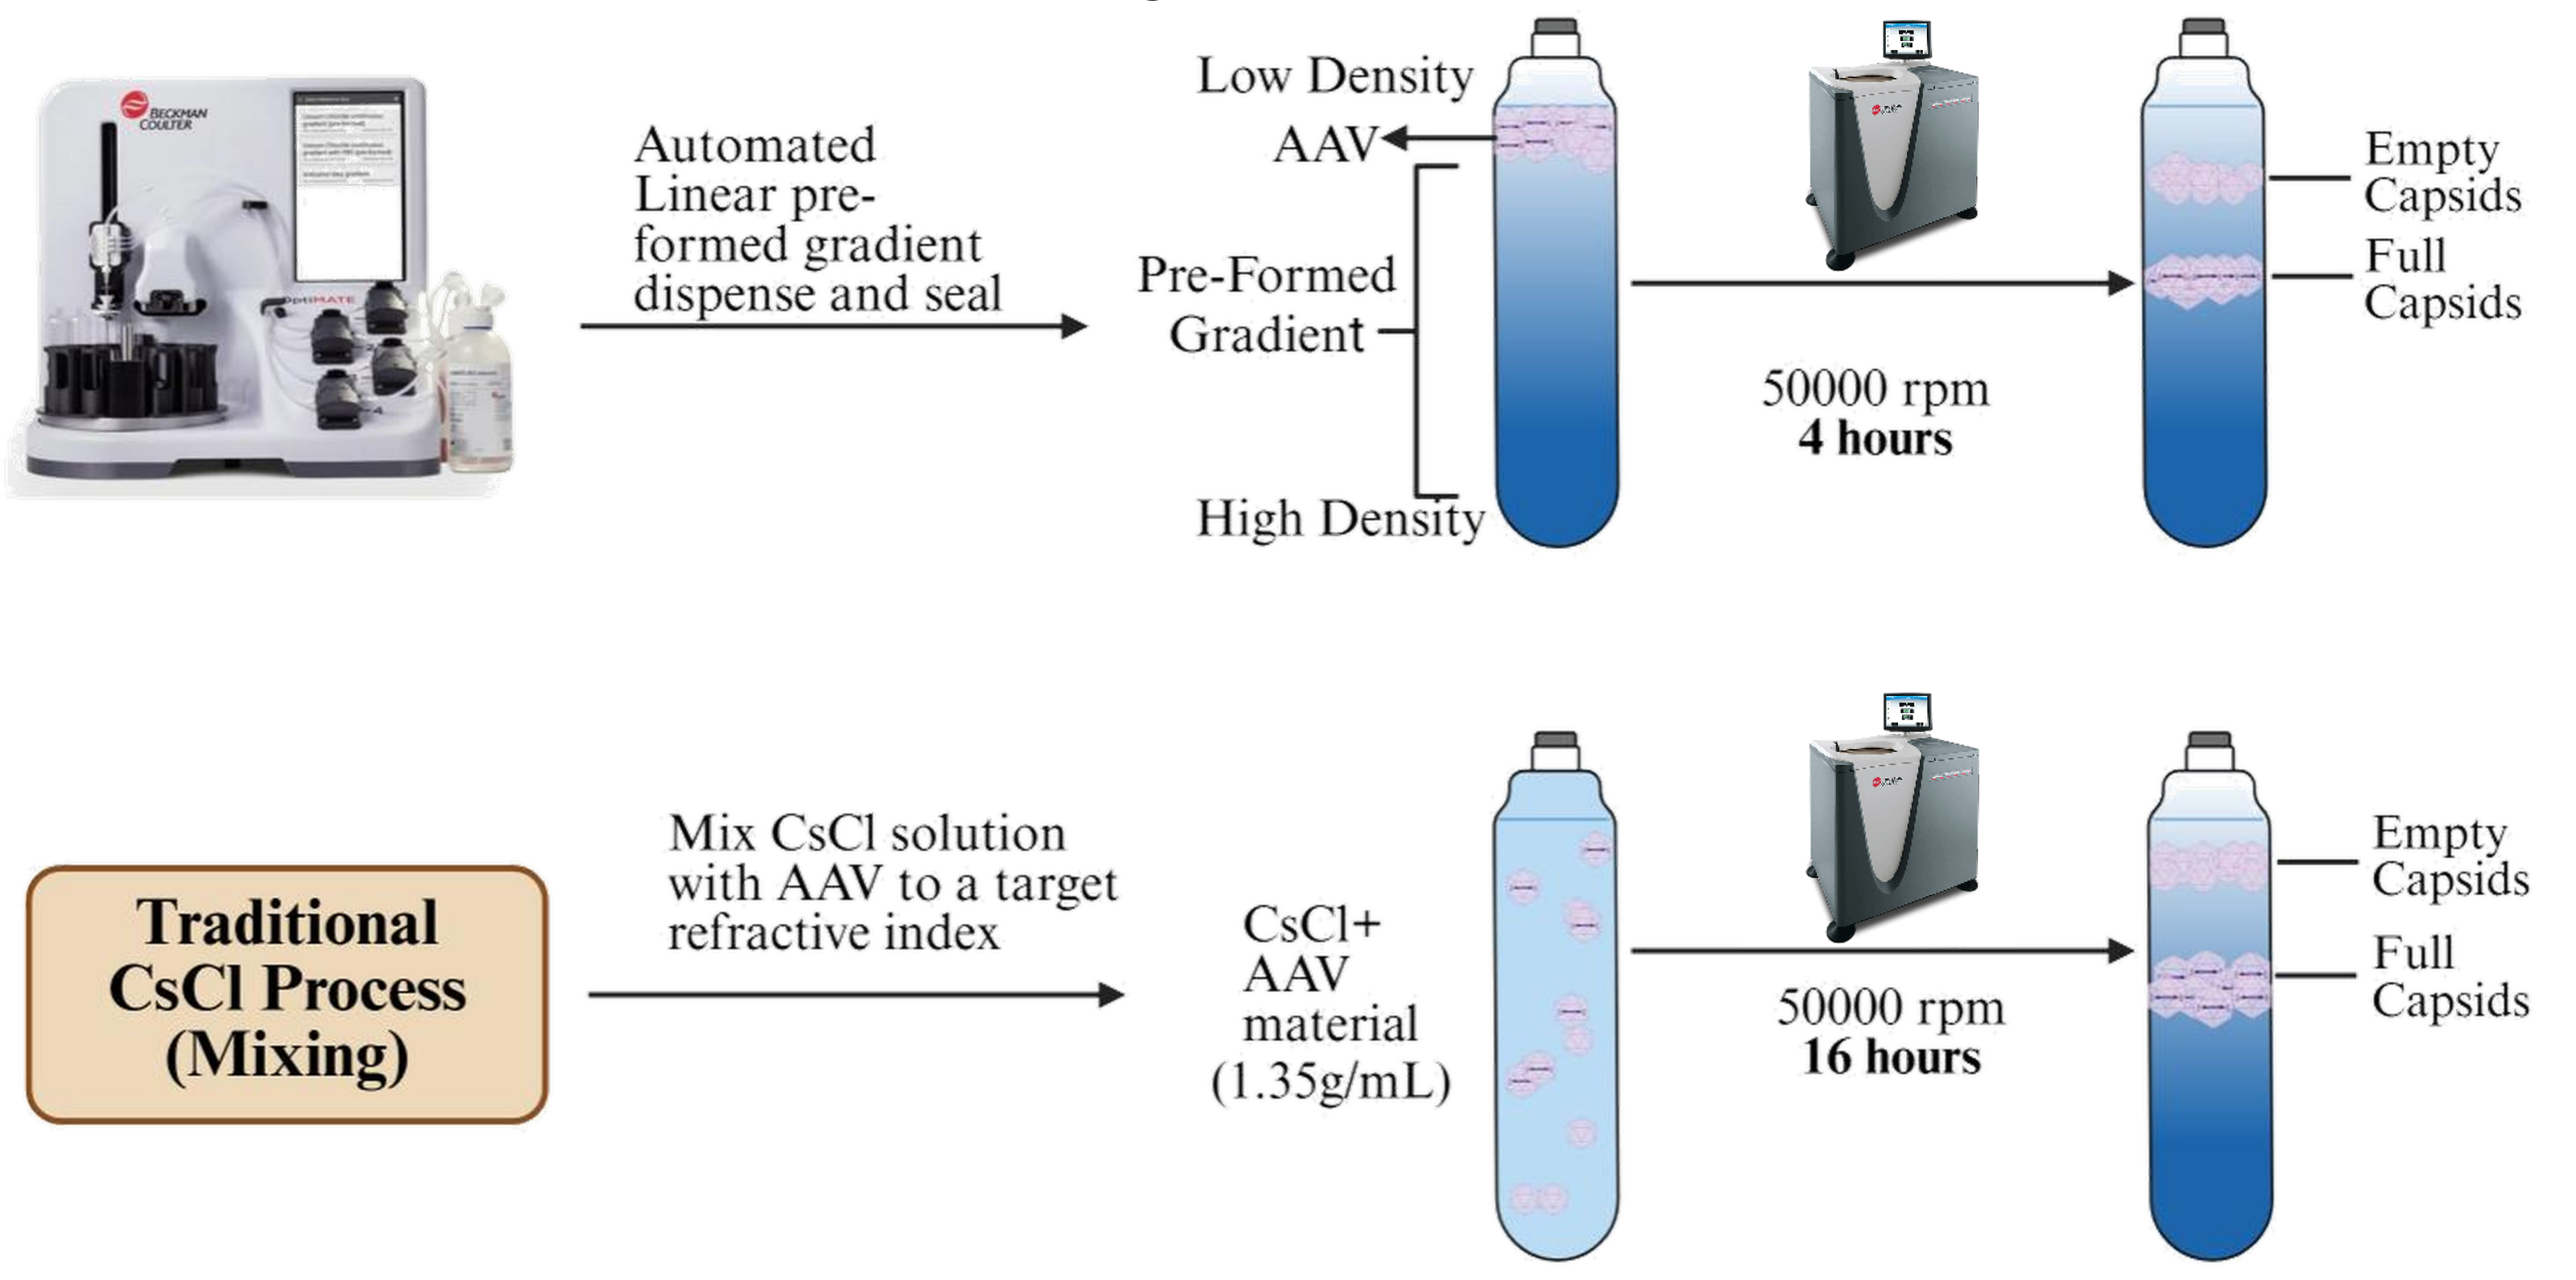
Automated Linear pre-formed gradient dispense and seal
Low Density
AAV
Pre-Formed Gradient
High Density
50000 rpm
4 hours
Empty Capsids
Full Capsids
Traditional CsCl Process (Mixing)
Mix CsCl solution with AAV to a target refractive index
CsCl+ AAV material (1.35g/mL)
50000 rpm
16 hours
Empty Capsids
Full Capsids

Figure 2. Comparison of CsCl Gradient Approaches. Pre-formed gradient (OptiMATE system) enables faster separation of full and empty capsids compared to self-formed gradient (Traditional), reducing spin time while maintaining resolution.

Parameters	Experiment 1 OptiMATE System Process	Experiment 2 Forge Platform Process (Traditional)
RPM	50,000	50,000
Temperature	17 °C	17 °C
Tubes	2x39.2 mL	2x39.2 mL
Refractive Index	1.3710–1.3730	1.3709
Neutralized Eluate (mL)	40.8 mL (20.4 mL each tube)	27 mL
Spin Time	**4 h**	**≥16 h**

Table 1. CsCl Run Parameters. Comparison between pre-formed and self-formed gradient process parameters

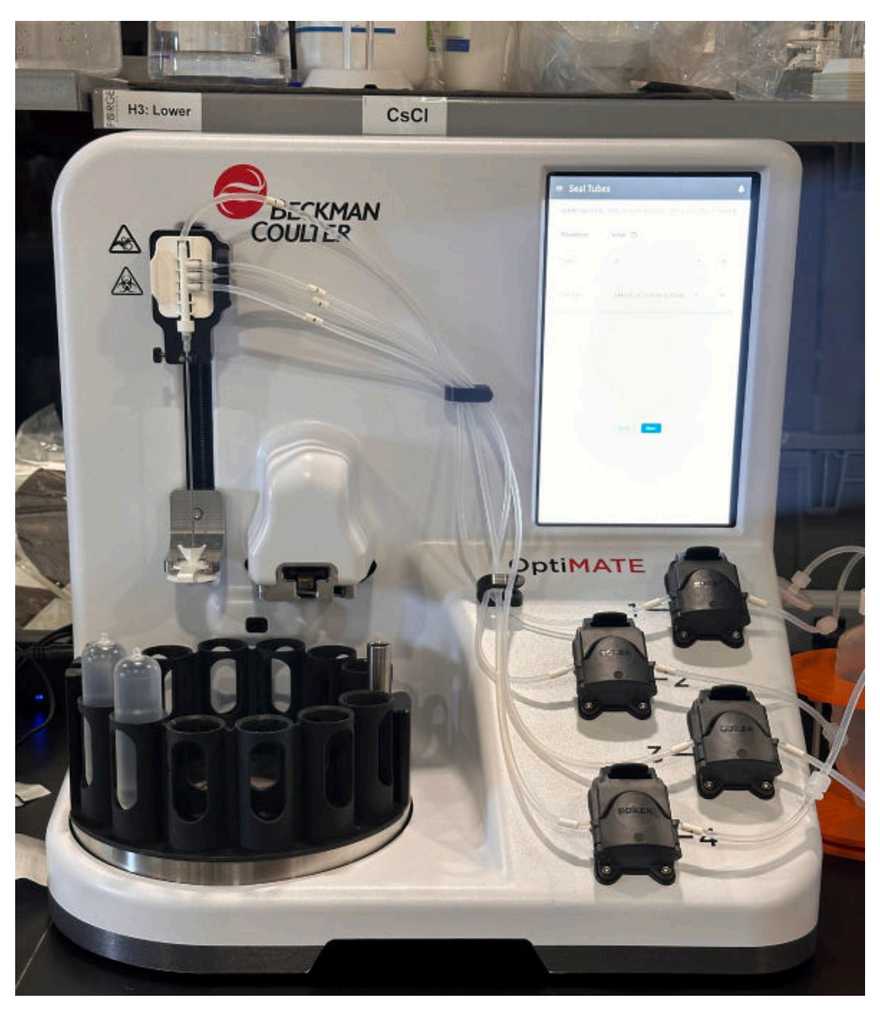

Figure 3. OptiMATE instrument setup. Ready to start CsCl tube filling and sealing

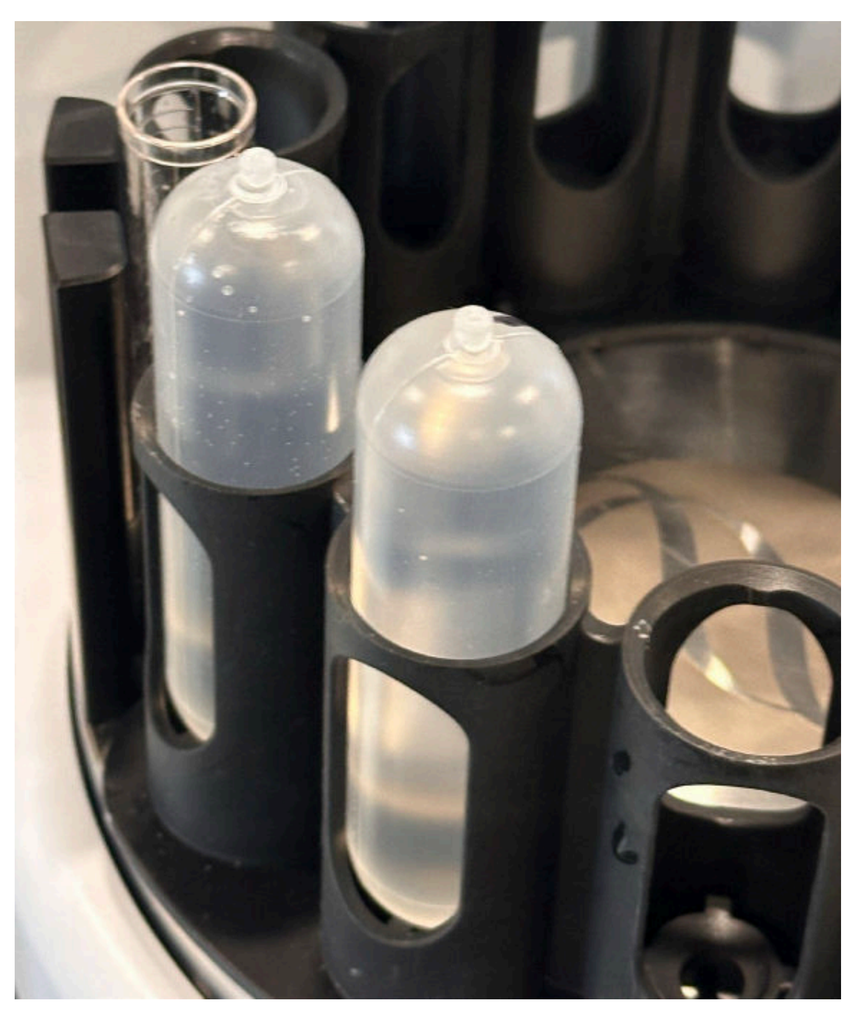

Figure 4. Tubes post-sealing using the OptiMATE system. Produced uniform and reliable tube seal

RESULTS

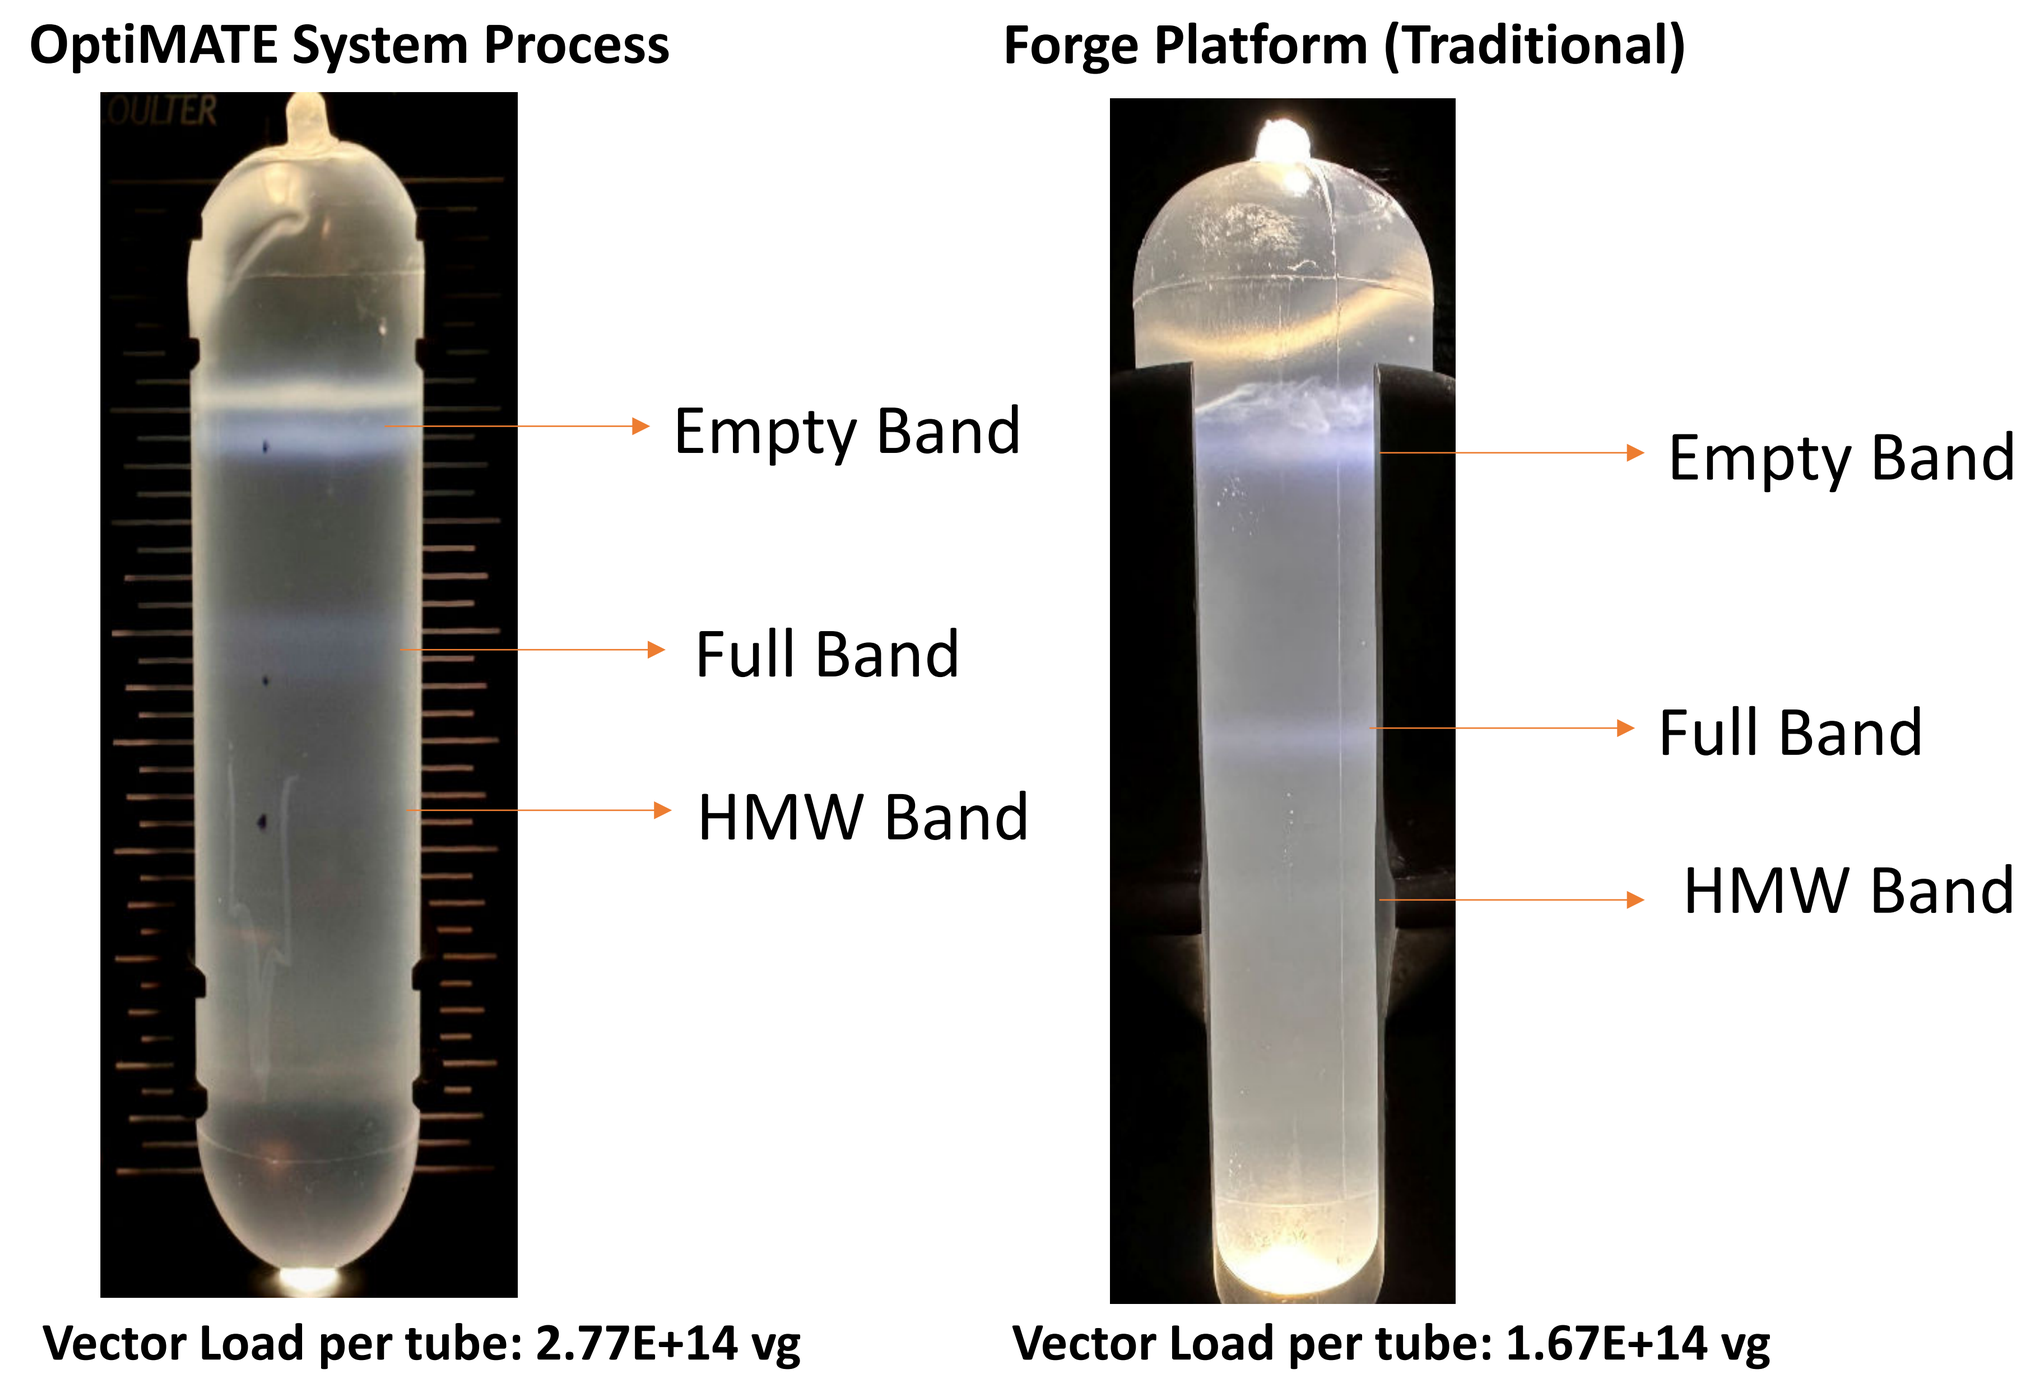
OptiMATE System Process
Forge Platform (Traditional)
Empty Band
Full Band
HMW Band
Empty Band
Full Band
HMW Band
Vector Load per tube: 2.77E+14 vg
Vector Load per tube: 1.67E+14 vg

Figure 5. CsCl Tube post ultracentrifugation spin. Three separate bands (Empty, Full and High Molecular Weight) were observed post-spin.

Condition	Sample	ddPCR Titer (vg/mL)	Yield (vg)	Step Recovery (%)
OptiMATE System Process	CsCl Load	1.36E+13	5.54E+14	**N/A**
OptiMATE System Process	**CsCl Full Band**	**3.23E+13**	**4.16E+14**	**75.09**
Forge Platform (Traditional)	CsCl Load	4.44E+12	3.33E+14	**N/A**
Forge Platform (Traditional)	**CsCl Full Band**	**1.93E+13**	**2.41E+14**	**72.37**

Table 2. Table summarizes ddPCR titers, yield, and step recoveries between the OptiMATE System and Forge Platform processes (Traditional).

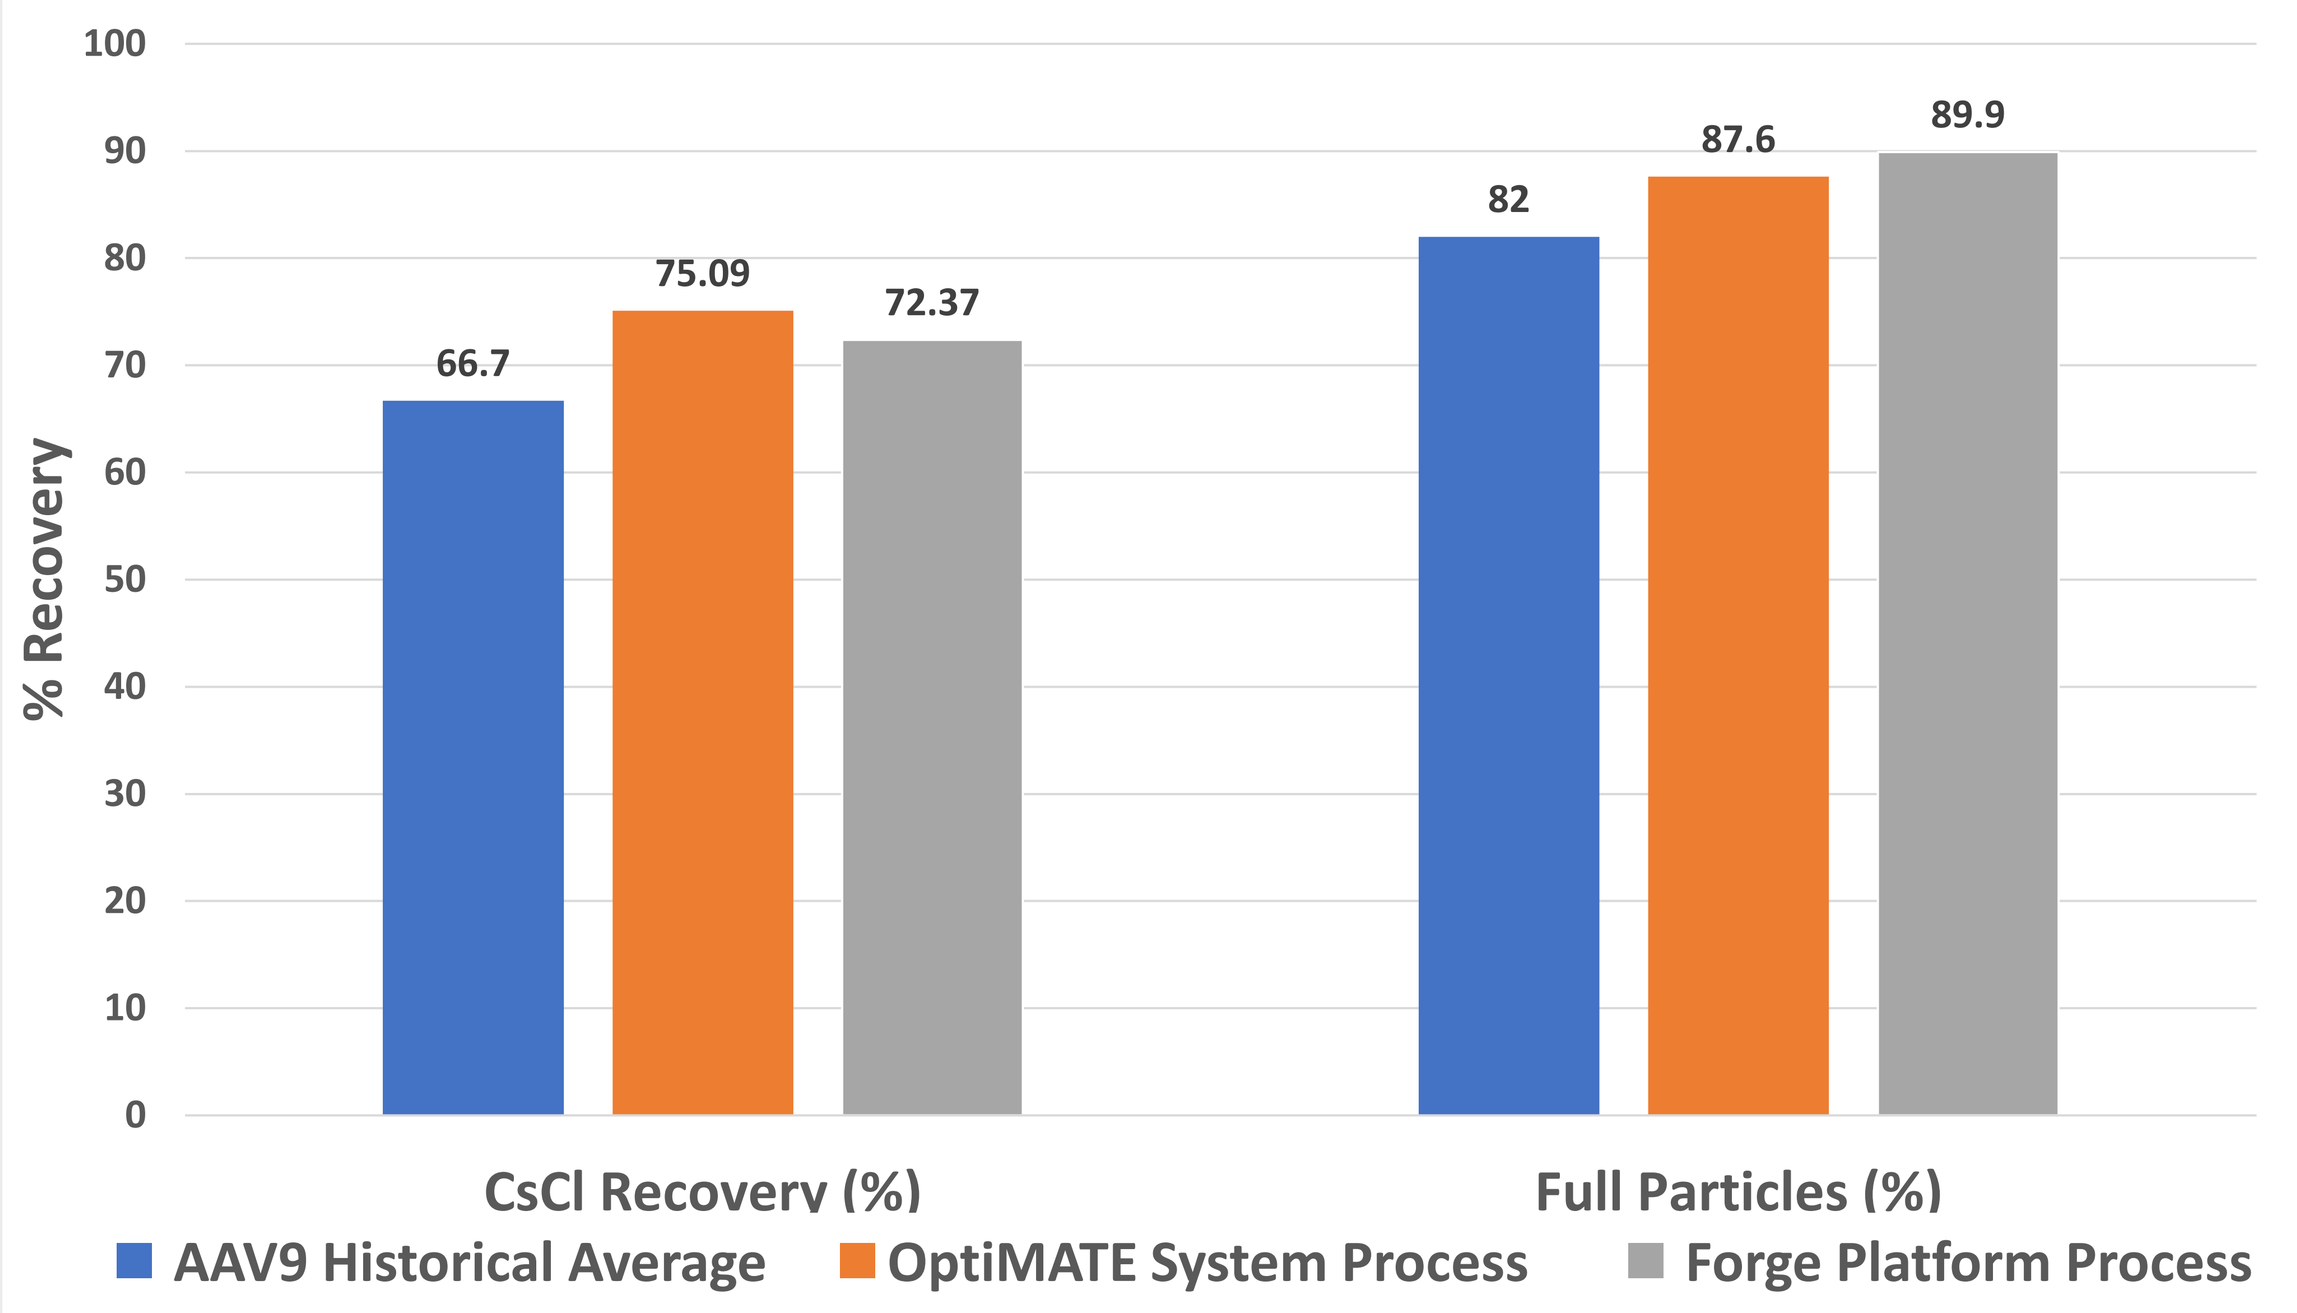
% Recovery
100
90
80
70
60
50
40
30
20
10
0
66.7
75.09
72.37
82
87.6
89.9
CsCl Recovery (%)
Full Particles (%)
AAV9 Historical Average
OptiMATE System Process
Forge Platform Process

Figure 6. Comparison of %Full band recovery and % Full Particles by AUC. CsCl recovery was comparable between OptiMATE System (75.09%) and Forge Platform (72.37%), with both being above historical averages (66.7%).% Full particles were also comparable (OptiMATE System 87.6%, Forge Platform 89.9%) relative to historical averages (82%).

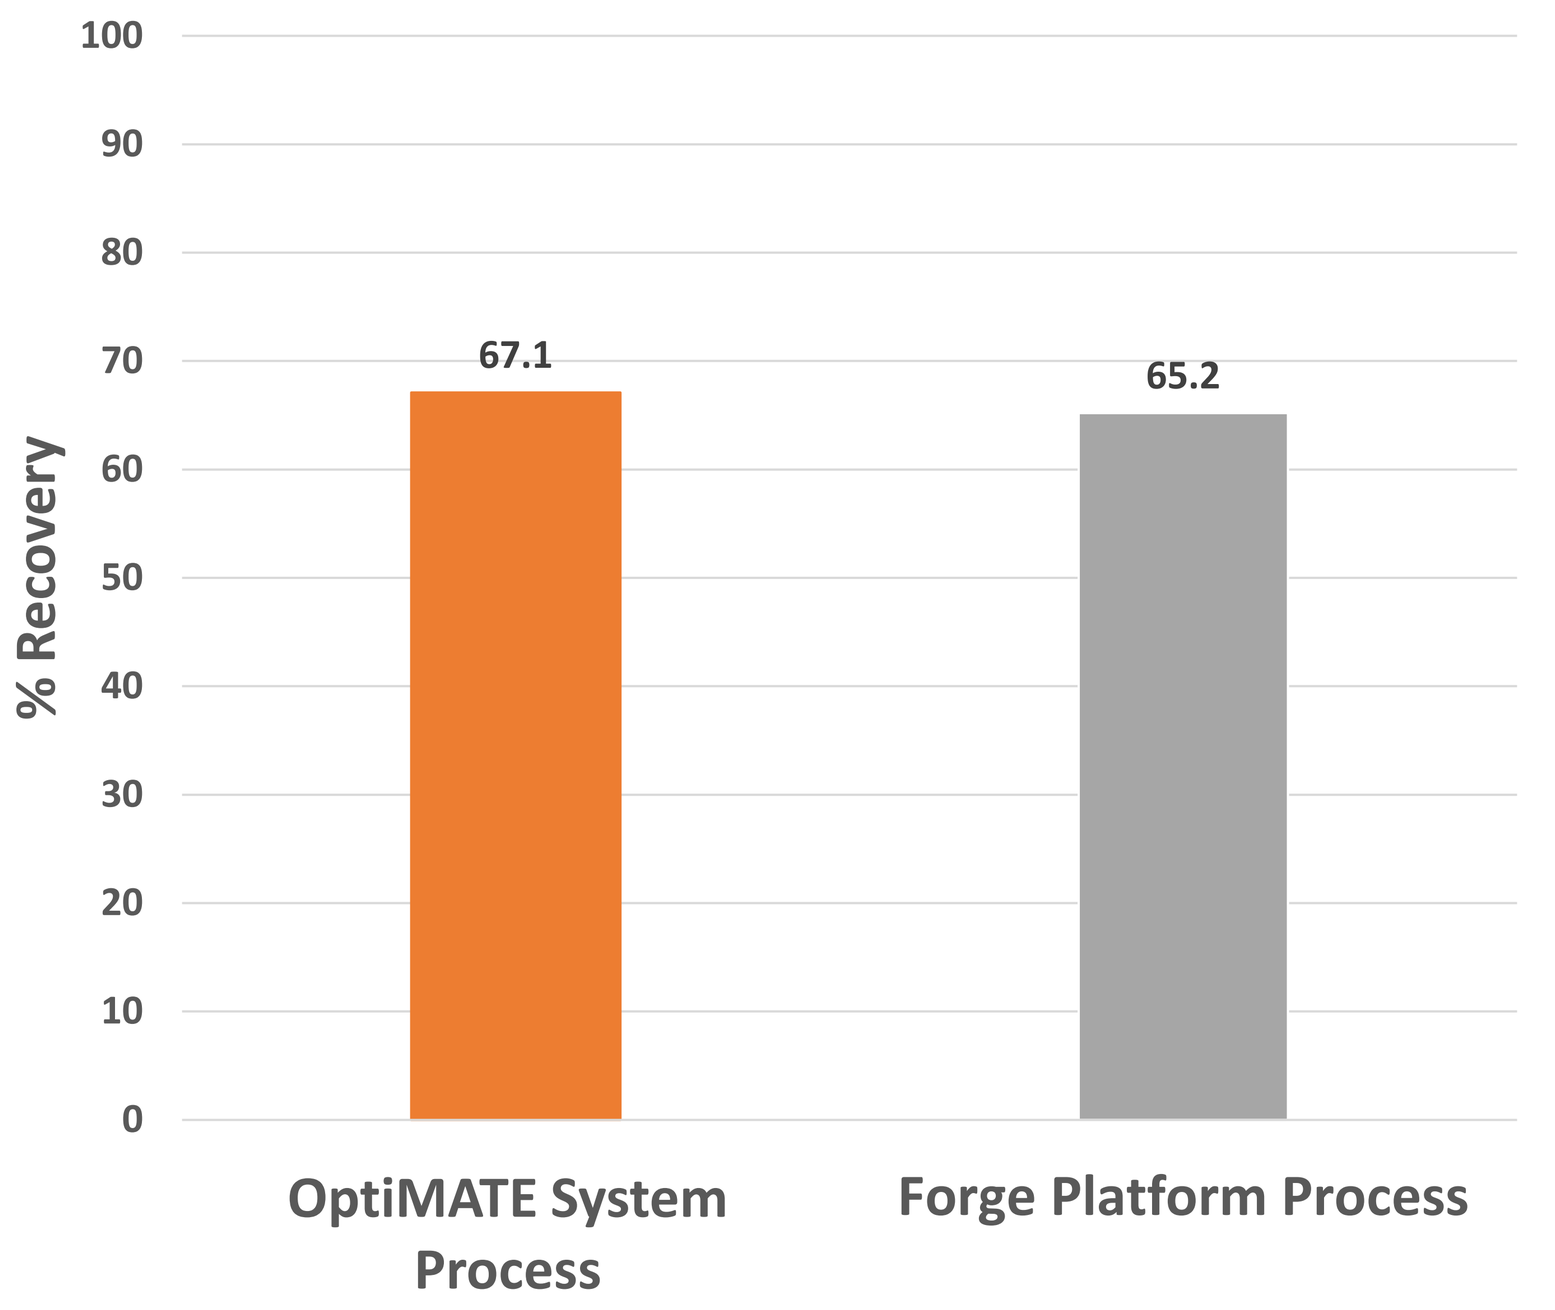
% Recovery
100
90
80
70
60
50
40
30
20
10
0
67.1
65.2
OptiMATE System Process
Forge Platform Process

Figure 7. Recovery from CsCl Load to Final Formulation. Recovery was 67.1% with OptiMATE System and 65.2% with Forge Platform, demonstrating comparable performance between the two processes.

OptiXTRACT Sample Recovery Aid

Eliminate Safety Concerns
Simple slide rail ensures tubes can be punctured without the need to hold the tube
Reduce Error & Improve Consistency
Graduations and a dimmable light allow for accurate, reproducible piercing and band extraction
Enhanced Flexibility
Supports multiple Luer-Lok™ syringes with customizable positions and is compatible with various tube sizes
Suitable for sterile operation
Small equipment footprint that fits easily inside a biosafety cabinet; sterile syringe adapters, perfect for single use

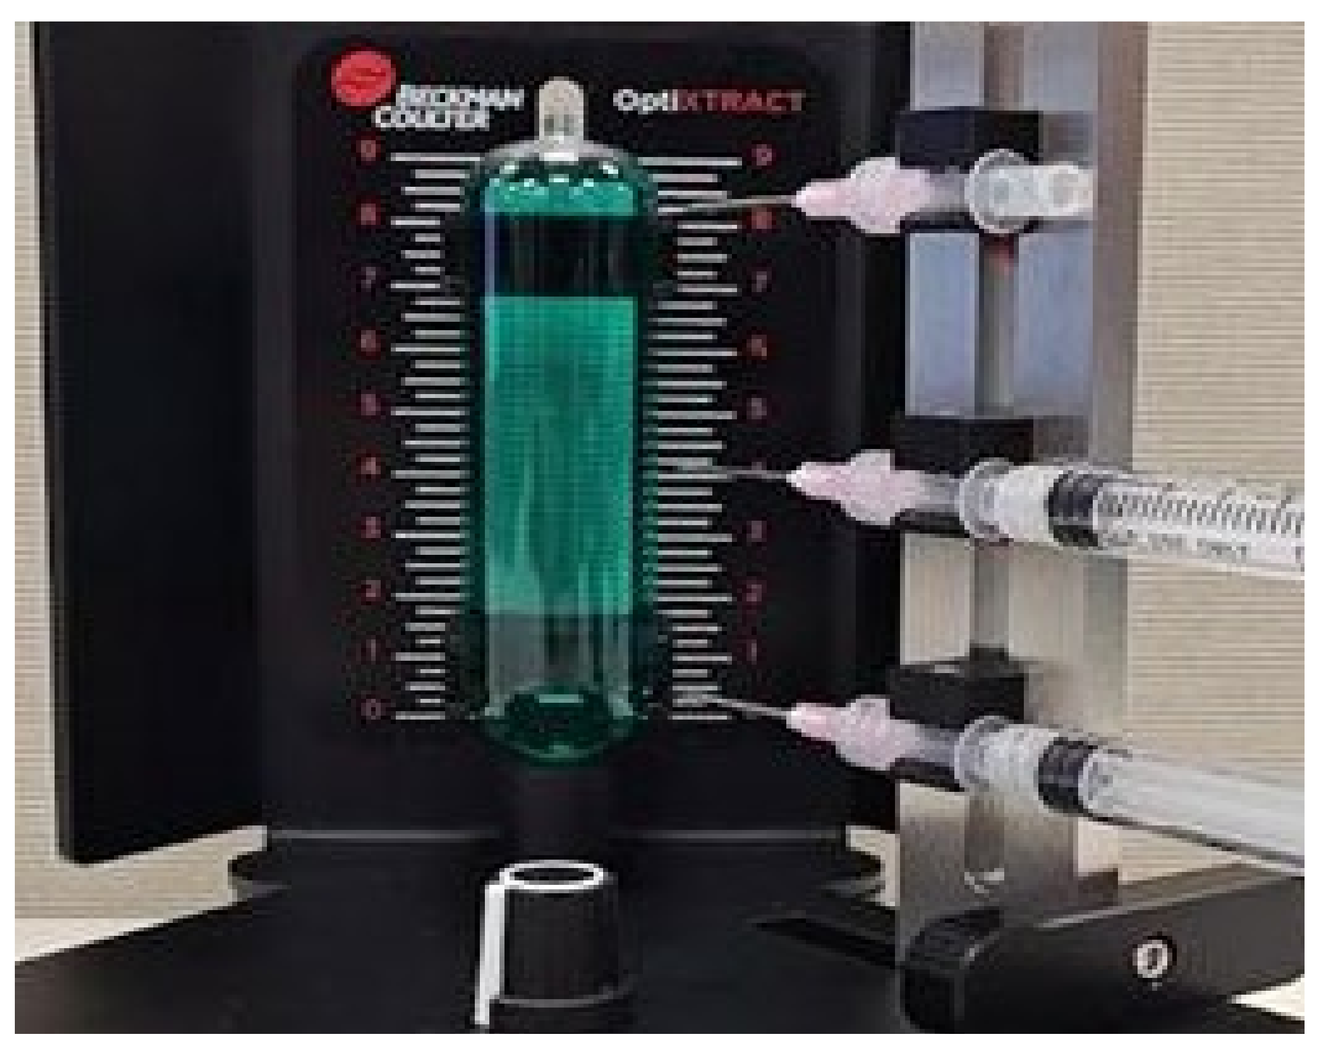
BECKMAN COULTER
OptiXTRACT

Key Takeaways

- The results from the gradient maker testing show **similar recoveries and quality attributes** to the Traditional Forge platform CsCl process.
- **Shorter processing times**, makes it possible to perform more spins per day.
- Preconfigured methods ensure **consistent and reproducible** results across different lots.
- Provides **uniform and reliable sealing** across tubes, removing operator-dependent variation.
- Eliminates bubble formation while filling the tubes. **Removes the need for balancing** the tubes.
- **The OptiXTRACT** extraction tool offers excellent stability for band collection, consistent pull based on distance and removes operator variability.

Conclusion

Overall, the results indicate that the outcomes using the automated gradient maker are comparable to the Forge platform process, both in terms of recovery and product quality, while offering significant time savings and reproducibility for large-scale operations.

References:
1. Zwi-Dantsis L et al.: Adeno-associated virus vectors: principles, practices, and prospects in gene therapy. Viruses 2025; 17(2): 239

Contact: Anto Nirmal Louis Babu,
Senior Associate Scientist, Forge Biologics
Email id: anirmal@forgebiologics.com

The OptiXTRACT Sample Recovery Aid is in development and performance characteristics are not validated.
For Research Use Only. Not for use in diagnostic procedures. The method described is for demonstration only and is not validated by Beckman Coulter. Beckman Coulter, the stylized logo, and the Beckman Coulter product and service marks mentioned herein are trademarks or registered trademarks of Beckman Coulter, Inc. in the United States and other countries. All other trademarks are the property of their respective owners. 2025-GBL-EN-108595-v1

AJINOMOTO BIO•PHARMA SERVICES